Automatic kV Selection for Radiation Dose Reduction in CT: How Does it Work and What Can it Do?

Michael R. Bruesewitz, R.T.(R), Lifeng Yu, Ph.D., Shuai Leng, Ph.D., Thomas J. Vrieze, R.T.(R),
Lingyun Chen, Ph.D., James M. Kofler, Ph.D., Joel G. Fletcher, M.D., Cynthia H. McCollough, Ph.D.

Department of Radiology, Mayo Clinic, Rochester, MN

Introduction

Concerns with the potential risk of cancer induction resulting from the radiation dose in CT exams have arisen with the drastically increased use of CT. Although the existence of such risk remains controversial for the level of radiation doses typically received in diagnostic CT, consensus is that patients should receive radiation dose as low as reasonably achievable (ALARA).

A commonly used method to reduce radiation dose is automatic exposure control (AEC), which automatically adapts the tube current in both angular and longitudinal directions according to patient attenuation to achieve predefined image quality.(1) Another important technique is to adjust tube potential (kV). Many researchers have studied this technique. A common critical finding in these studies was the appropriateness of using lower kV is highly dependent on patient size and diagnostic task.(2) For smaller patients and some types of contrast-enhanced studies such as CT angiography (CTA), the dose reduction can be 50% or even higher. But for bigger patient sizes and other exam types, the image quality may become unacceptable if using the lower kV even without any radiation dose reduction. Selection of an optimal kV should take into account both the patient size and diagnostic task. This is not a trivial task in clinical practice and demands a quantitative approach that can automatically determine the optimal kV and the amount of radiation dose reduction. Automatic selection of kV should be incorporated into the AEC in addition to the automatic tube current modulation in order to provide a convenient approach to optimizing the dose efficiency of scanning technique without much user interactions.(3)

In this exhibit, we first describe why the kV needs to be optimized and introduce the basic principles of automatic selection of kV. Then we summarize recent development on automatic techniques to select the most dose-efficient kV. Some important considerations in clinical practice are also discussed.

Why kV Needs to be Optimized

Contrast

Many CT exams involve the use of iodinated contrast media. The different energy dependence of the linear attenuation coefficients for iodine and water leads to different CT numbers of iodine at different kV. The increase of the CT number of iodine at lower kV provides more iodine signal and hence improves the conspicuity of hyper-vascular or hypo-vascular pathologies.

Figure 1 displays CT images of three water phantoms scanned using four different kVs available on a 128-slice scanner (Definition Flash, Siemens Healthcare). The lateral widths of the three phantoms were 25 cm, 35 cm, and 45 cm, representing typical attenuation levels for a small, average, and large sized adult, respectively. For each phantom size, the scanning technique of quality reference mAs at each kV was adjusted so that the radiation output, represented in terms of CTDIvol, was matched for the four kVs (25 cm: 6.6 mGy; 35 cm, 15.3 mGy; 45 cm, 37.0 mGy). AEC was turned on. Several different contrast materials were placed inside the water to allow measurement of contrast.

Figure 2 plots the contrast of iodine (the sample with an iodine concentration of 6.9 mg/cc, see arrows on 120 kV images) at the four kVs. On average, the iodine contrast of 80 kV was about 70% and 100% higher than that of 120 kV and 140 kV, and the iodine contrast of 100 kV was about 25% and 50% higher than that of 120 kV and 140 kV, respectively. The increase of iodine contrast at lower kV varies with the phantom size due to the beam hardening effect.

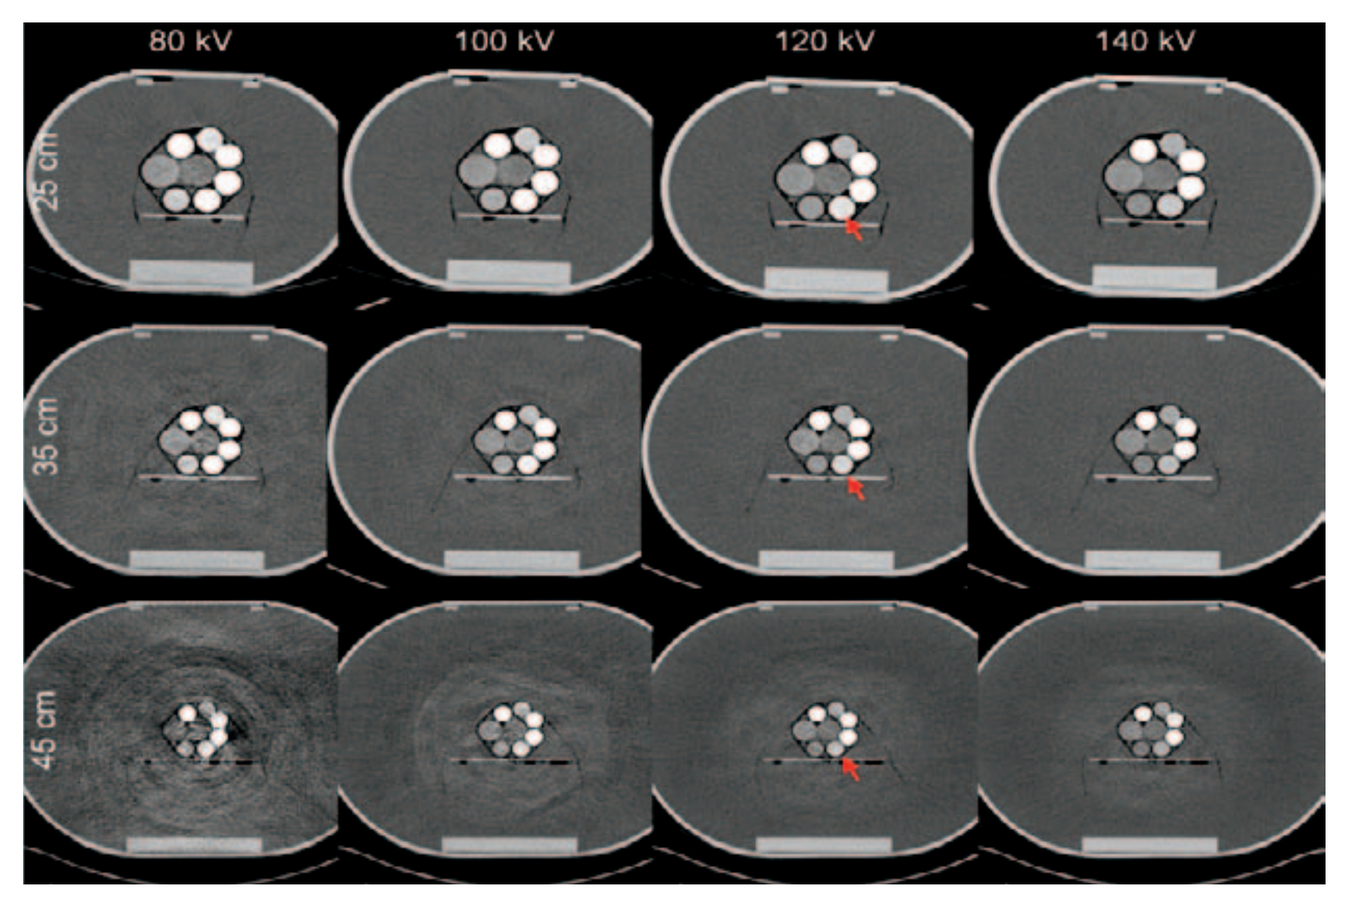
80 kV
100 kV
120 kV
140 kV
25 cm
35 cm
45 cm

Figure 1. CT images of three water phantoms scanned with four kVs at matched CTDIvol. The phantom lateral width was 25 cm, 35 cm, 45 cm. For each phantom size, the prescribed CTDIvol was matched for the four kVs (25 cm: 6.6 mGy; 35 cm, 15.3 mGy; 45 cm, 37.0 mGy).

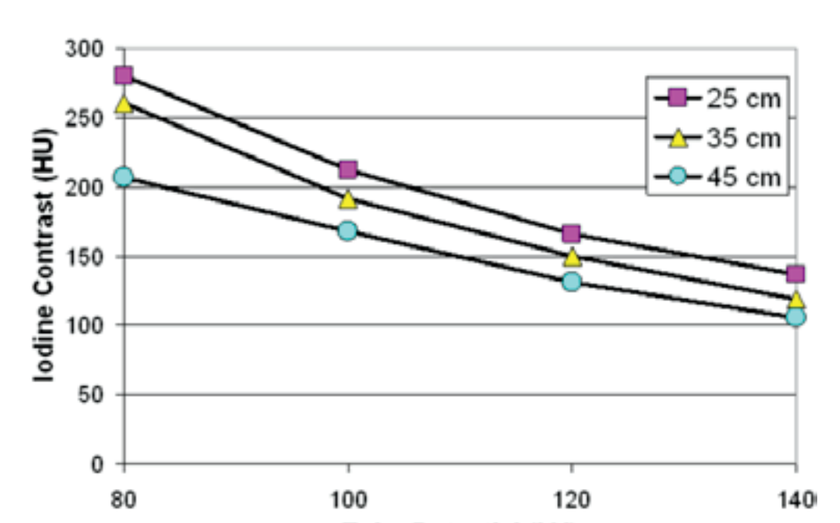
Iodine Contrast (HU)
Tube Potential (kV)
25 cm
35 cm
45 cm

Figure 2. The change of iodine contrast with the kV for different phantom sizes. For each phantom size, the CTDIvol was held constant as kV varied.

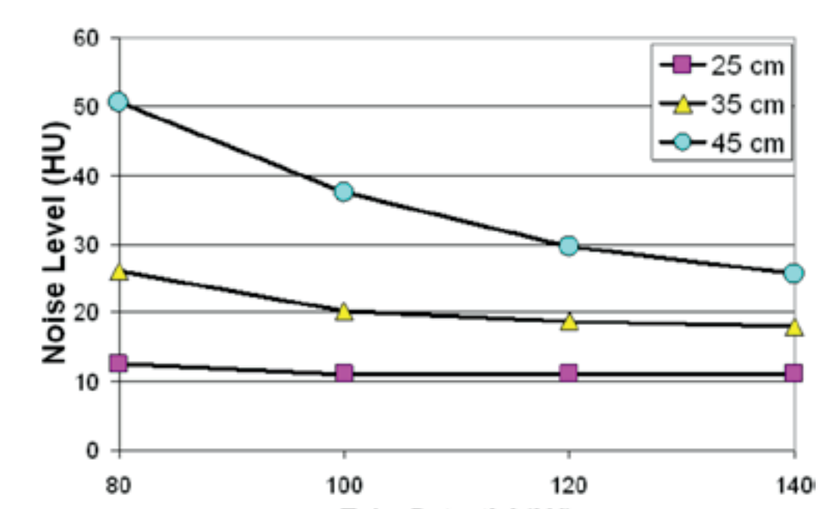
Noise Level (HU)
Tube Potential (kV)
25 cm
35 cm
45 cm

Figure 3. The change of noise level with the kV for different phantom sizes. For each phantom size, the CTDIvol was held constant as kV varied.

Noise

Noise is another important factor that greatly influences the image quality. The noise level (expressed as the standard deviation of the CT number) is affected by kV and patient size. Noise spatial correlation and higher-order statistics also contribute significantly to the image quality, but they remain similar at different kV provided that other factors such as reconstruction algorithms are the same.

Figure 3 shows the noise level measured on the three phantom sizes at each of the four kVs (data from the same measurement as in **Figure 1**). For the 25 cm phantom size, noise level is similar at 100 kV, 120 kV, and 140 kV and there is a slight increase at 80 kV. For the 35 cm and 45 cm phantom, noise increases substantially on the 80 kV images. In addition, significant photon-starvation artifacts appears in the 80 kV images of the large phantom, due to the decreased penetrating capability of the lower energy photons and the electronic noise.

Figure 4 shows a clinical example demonstrating the increased noise at 80 kV compared with 120 kV at the same CTDIvol.

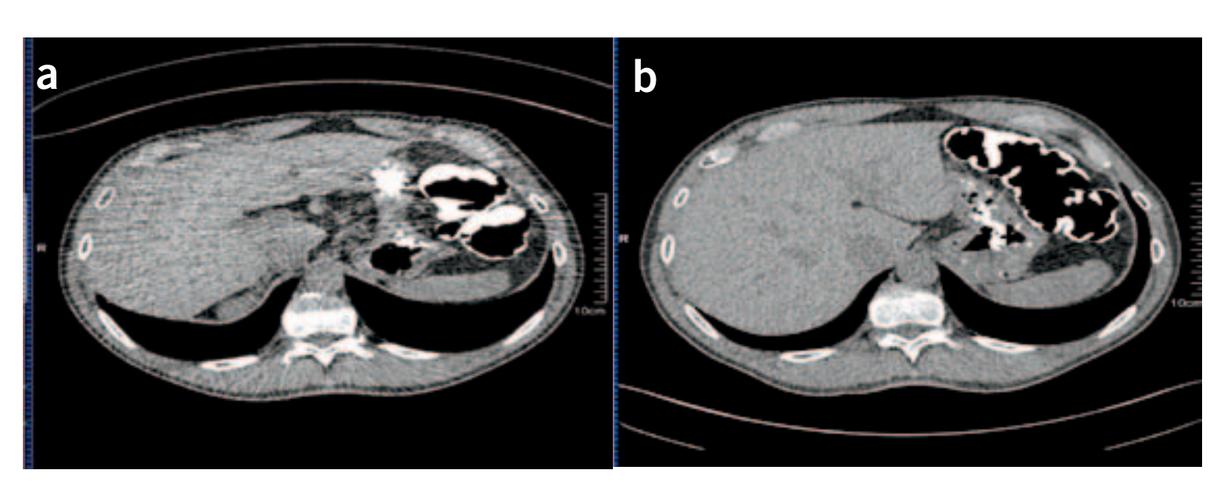
a
b

Figure 4. Clinical exam of increased noise (a) 80kV CTDIvol=6.4 mGy (b) 120 kV CTDIvol=6.5mGy

Contrast to Noise Ration (CNR)

CNR is typically used to represent the combined effect of contrast and image noise – both of which are important image quality metrics. CNR cannot be used to quantify the absolute image quality of an image as it does not take into account the effect of system spatial resolution, noise texture, and object size. However, if all other factors are the same, then CNR can serve as a relative measure to compare image quality. CNR is often expressed in terms of iodine contrast divided by noise in the background structures as iodine is the most widely used contrast material in CT.

Figure 5 shows the iodine CNR at each of the four kVs for the three phantoms. The improvement of iodine CNR for the 25 cm phantom at lower kV is very significant (almost doubled). The amount of increase of iodine CNR decreases for bigger size phantoms because of the combined effect of the increased noise and the increased contrast. The iodine CNR at 80 kV for the 35 cm phantom still increases, but it drops slightly for the 45 cm phantom. Based on **Figure 5**, it appears that 80 kV or 100 kV images are similar to or better than 120 kV images in terms of iodine CNR. However, the actual image quality degradation of lower kV for large phantom actually cannot be fully characterized by the iodine CNR. As shown in **Figure 1** there were very severe photon starvation artifacts in the 80 kV image for the 45 cm phantom. Because of this reason, lower kV should not be used for large patients.

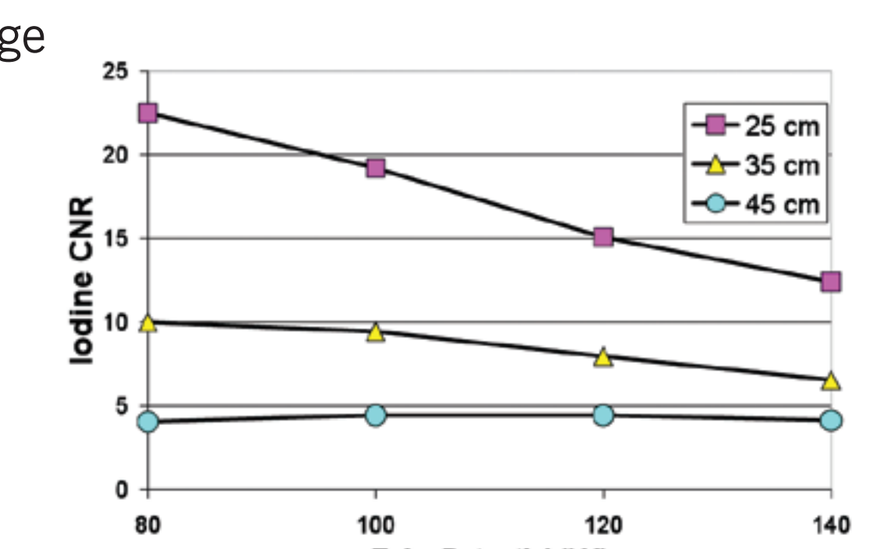
Iodine CNR
Tube Potential (kV)
25 cm
35 cm
45 cm

Figure 5. The change of iodine contrast to noise ratio (CNR) with the kV for different phantom sizes. For each phantom size, the CTDIvol was held constant as kV varied.

Basic Principles of Optimal kV Selection

Based on the relationship between contrast, noise, and contrast to noise ratio described previously, the appropriateness of using lower-kV is highly dependent on patient size and diagnostic task.

The optimal kV is the kV that uses the minimum radiation dose to achieve the desired image quality. In order to select the most-dose efficient kV and quantify the amount of dose reduction, one should set up a "desired image quality" using a certain image quality metric.(4)

The desired image quaility can be defined using image quality metrics such as noise or iodine CNR. The amount of radiation dose required to match the desired image quality, quantified with the selected image quality metric, at each kV can be calculated and subsequently the most dose-efficient kV can be selected.

Radiation Dose Reduction if Iodine CNR is Matched

With the increased iodine CNR at lower kV, one could reduce the radiation dose and achieve similar or improved iodine CNR relative to the more commonly used 120 kV. **Figure 6** displays the relative CTDIvol at each kV if the same iodine CNR is to be achieved. For the 25 cm phantom, the CTDIvol needed for identical CNR relative to that at 120 kV is 46% at 80 kV and 62% at 100 kV. The potential for dose reduction decreases with the increase of phantom size. For the 35 cm phantom, 64% at 80 kV and 72% at 100 kV are needed. For the 45 cm, one need 18% more dose at 80 kV than at 120 kV in order to match the iodine CNR.

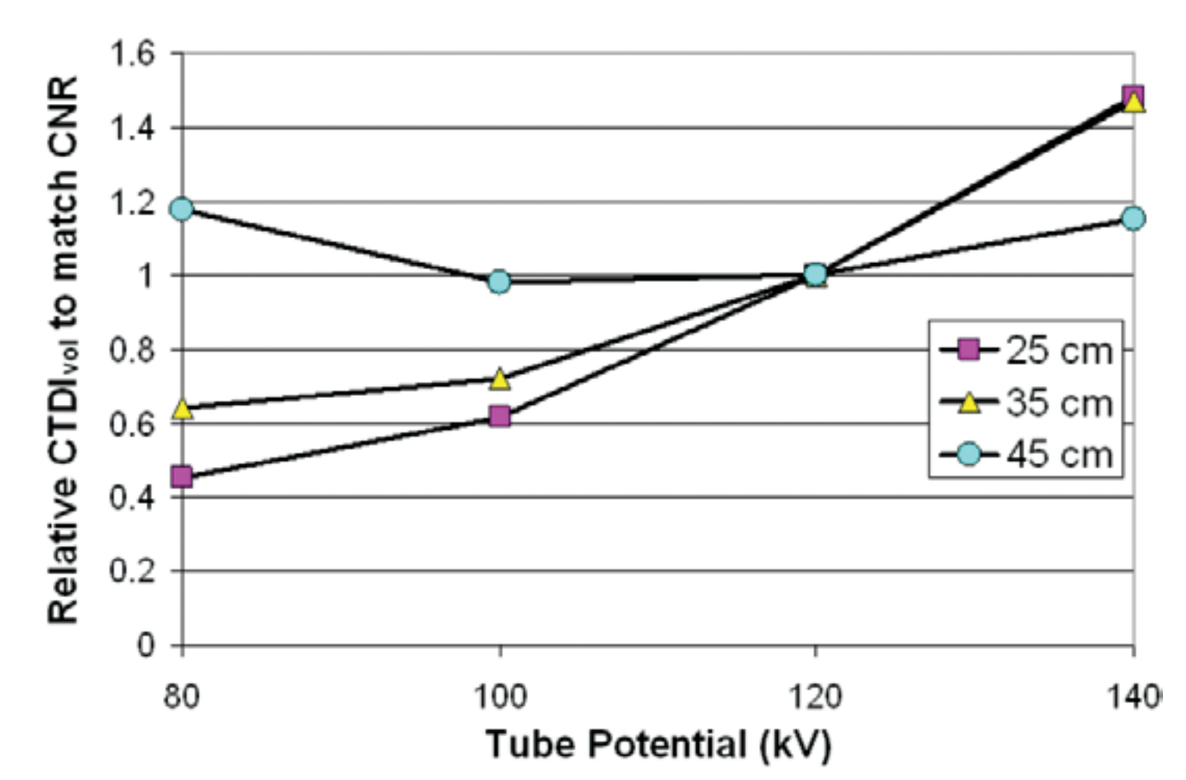
Relative CTDI$_{vol}$ to match CNR
Tube Potential (kV)
25 cm
35 cm
45 cm

Figure 6. The relative radiation output required at each kV to obtain the same iodine contrast to noise ratio (CNR) for the three phantoms.

Radiation Dose Reduction if Noise is Matched

Based on the measurements of noise at equivalent doses, the relative dose that is required at each kV in order to achieve the same noise level can be estimated. **Figure 7** clearly demonstrates that if the noise of a 120 kV image is matched, the potential for dose reduction at lower kV is very limited or non-existent. Even for the 25 cm phantom that represents the attenuation of a very small adult, a 29% dose increase is required at 80 kV in order to match the noise. For the 35 cm phantom representing the attenuation of a medium-sized adult required a 94% dose increase at 80 kV and 18% dose increase in 100 kV in order to compensate for the increased noise. And the increase of dose for 45 cm phantom at lower kV is even more dramatic.

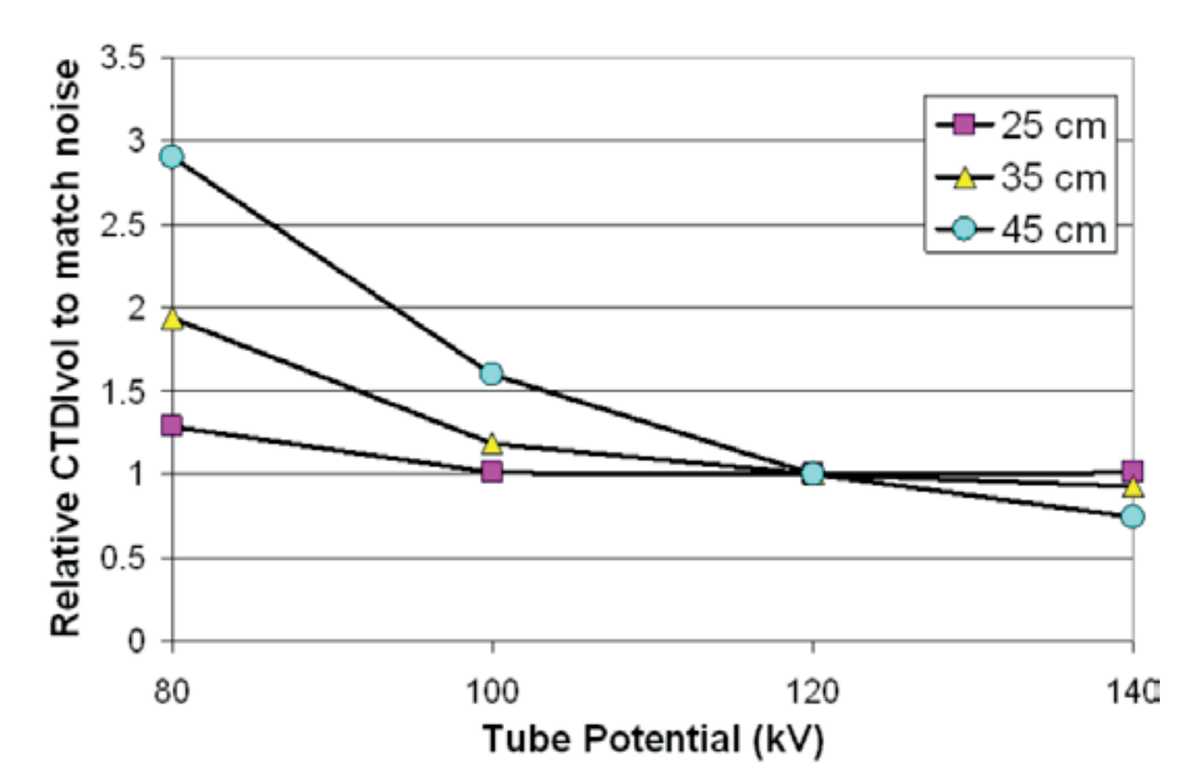
Relative CTDIvol to match noise
Tube Potential (kV)
25 cm
35 cm
45 cm

Figure 7. The relative radiation output required at each kV to obtain the same noise level for the three phantoms.

Radiation Dose Reduction When both Iodine CNR and Noise are Incorporated

One can see that the selection of the most dose-efficient kV and the estimate of the amount of dose reduction in CT are highly dependent on the image quality metric that is used for matching at different kVs. The image quality metric is subsequently determined by the clinical task performed. When the task only involves the evaluation of highly iodine-enhanced vessels or structures, iodine CNR may be an appropriate image quality metric to use. If the diagnostic task involves evaluation of non-enhanced soft tissue structures, then matching noise is more appropriate and the dose reduction is quite restricted using a lower kV. Many diagnostic tasks, such as routine contrast-enhanced abdomen/pelvis exams, are somewhere between these two scenarios. Lower kV brings some benefit on the contrast enhancement of iodine, but the noise cannot be too high. A scheme that can utilize the benefit of the contrast enhancement at lower kV but also can control the noise level is necessary to accommodate different diagnostic tasks. Therefore, an image quality index that can allow flexible adjustment between matching iodine CNR and matching noise appears to be attractive to determine the most dose-efficient kV.(5)

A General Strategy for Optimal kV Selection

To provide the flexibility between matching noise and matching iodine CNR, a novel image quality index, "noise-constrained iodine contrast to noise ratio (NC_iCNR)", was proposed to quantify the different levels of image quality required by different clinical applications for a reference dose level and kV.(5) This quality index requires that iodine CNR and noise in the new settings of kV and dose level satisfy the following two conditions:
$CNR \geq CNR_{ref}$ & $\sigma \leq \alpha\sigma_{ref}$.

Where CNR_{ref} and σ_{ref} denotes the iodine CNR and the noise level obtained in a reference scanning technique (e.g., a reference kV and mAs), respectively; α is a coefficient that specifies the level of noise constraint, which can be adjusted according to the diagnostic task. Maintaining a constant noise (α=1) or iodine CNR (α>2) are two special cases of this general image quality index.

Table I displays the optimal kV for seven different abdominal phantom sizes (in terms of lateral width) at five different noise constraint settings using the general strategy described above. The recommended noise constraint parameters for different exam types are also listed.

Table I. Optimal kV for different phantom sizes in abdominal CT exams when different noise constraints are applied.

Noise Constraint	Recommended exam types	25cm	30cm	35cm	40cm	45cm	50cm	55cm
Very weak α=1.00	Routine non-contrast exams	120	120	120	120	120	120	140
Weak α=1.15	Liver, pancreas exams	100	100	100	120	120	120	140
Average α=1.25	Routine contrast-enhanced exams	100	100	100	100	120	120	140
Strong α=1.50	CTE, CTU, stone, or some CTA exams	80	80	100	100	100	120	140
Very Strong α=2.00	CTA only involving large vessels	80	80	80	100	100	120	140

Clinical Implementation of Optimal kV

Clinical implementation of optimal kV selection is a complicated task. There are two methods: one is to implement a manual kV-mAs technique chart, the other is to implement a software tool on the scanner that can automatically select the optimal kV.

Manual kV – mAs Technique Chart

A convenient way to implement optimal kV is to use a patient weight or size-based kV-mAs chart, which specifies the kV, tube current (or mAs or effective mAs or reference mAs) for different patient weight or size ranges. The selection of the kV and the mAs level can be based on empirical evaluation or quantitative measurement on phantoms.

Table II provides two example kV-mAs charts implemented on a 128-slice scanner according to the phantom results and the general strategy for optimal kV selection. Note that the relative CTDIvol is beyond the reduction of radiation dose allowed by CAREDose4D, the automatic exposure control (AEC) software on Siemens CT scanners. The further dose reduction beyond AEC for smaller patients was enabled by the use of lower kV (80 kV or 100 kV). QRM represents quality reference mAs.

Table II. Example kV-mAs technique charts for (a) abdominal CTA exams, implemented on a 128-slice scanner (Definition Flash, Siemens Healthcare) and (b) contrast-enhanced routine abdomen/pelvis exams.

a

Patient lateral width (cm) – mid liver	Optimal kV	QRM	pitch	Relative $CTDI_{vol}$ (beyond CAREDose 4D)
<33	80	440	0.6	0.50
33-43	100	300	0.6	0.75
44-53	120	250	0.6	1.00
>53	140	170	0.6	1.00

b

Patient lateral width (cm) – mid liver	Optimal kV	QRM	pitch	Relative $CTDI_{vol}$ (beyond CAREDose 4D)
<30	80	580	0.5	0.70
30-40	100	330	0.8	0.85
41-50	120	240	0.8	1.00
>50	140	165	0.8	1.00

An Automatic kV selection Tool Implemented on the Scanner

An obvious disadvantage of the manual kV-mAs technique chart is its approximate determination of the patient attenuation level. Patient lateral width or other measures (weight, perimeter, etc) measured by technologists based on the scout or topogram is not accurate enough to represent the true patient attenuation level in the scan range. In addition, using the manual chart like in **Table II**, one has to prescribe a fixed amount of dose reduction for certain patient size range (e.g., 30 – 40 cm), which should have been gradually varying based on the patient attenuation level. The manual selection of the techniques may also be susceptible to additional human errors. Therefore, the selection of the optimal kV should be implemented on the CT scanner so that the software can automatically recommend the optimal kV and the reduced dose on each individual patient and each specific diagnostic task. The same general strategy as described above can be implemented in the automatic software.

One such software tool was recently developed by one of the major CT vendors (CAREkV, Siemens Healthcare). An example of using this software to select the optimal kV and to prescribe the dose-reduced technique is provided in **Figure 8**. The reference technique was at 120 kV and 250 quality reference mAs. A strength setting was configured for the exam through a slider bar, as shown in the user interface, which corresponds to a contrast gain constraint setting, equivalent to the noise constraint described in the general strategy. The software automatically determines the optimal kV and the dose-reduced technique. **Figure 9** explains different strength settings for the automatic kV selection software.

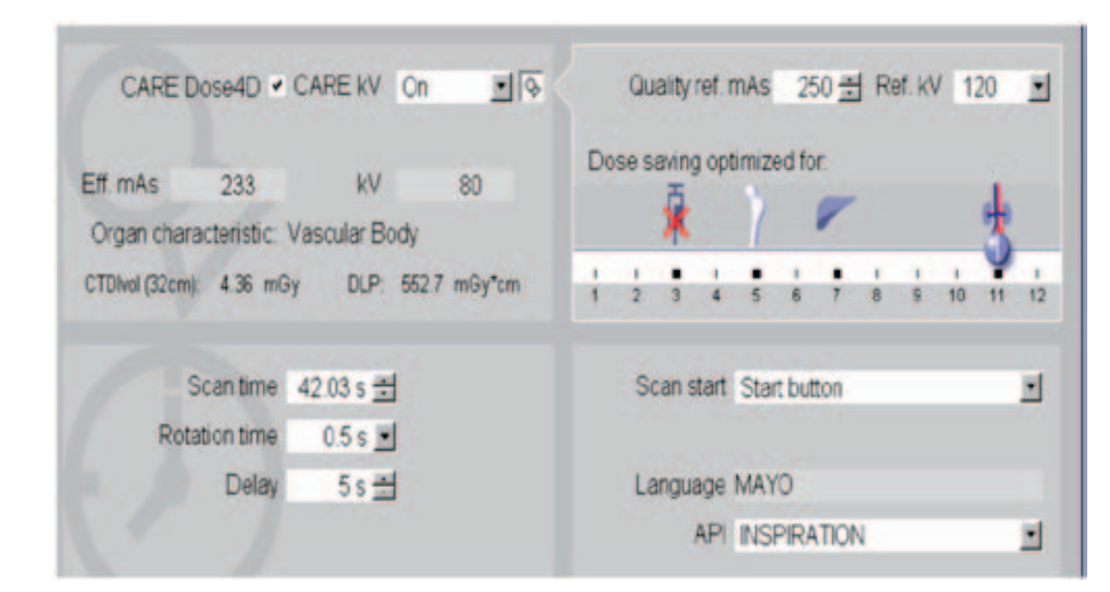
CARE Dose4D CARE kV On
Quality ref. mAs 250 Ref. kV 120
Eff. mAs 233 kV 80
Organ characteristic: Vascular Body
CTDIvol (32cm) 4.36 mGy DLP 552.7 mGy*cm
Dose saving optimized for
Scan time 42.03 s
Rotation time 0.5 s
Delay 5 s
Scan start Start button
Language MAYO
API INSPIRATION

Figure 8. Example of using an automatic software to select the optimal kV and to prescribe the dose-reduced technique (CAREkV, Siemens Healthcare). The reference technique was at 120 kV and 250 quality reference mAs. The slider bar position, which corresponds to a strength setting, was at 11. 80 kV was identified as the optimal kV.

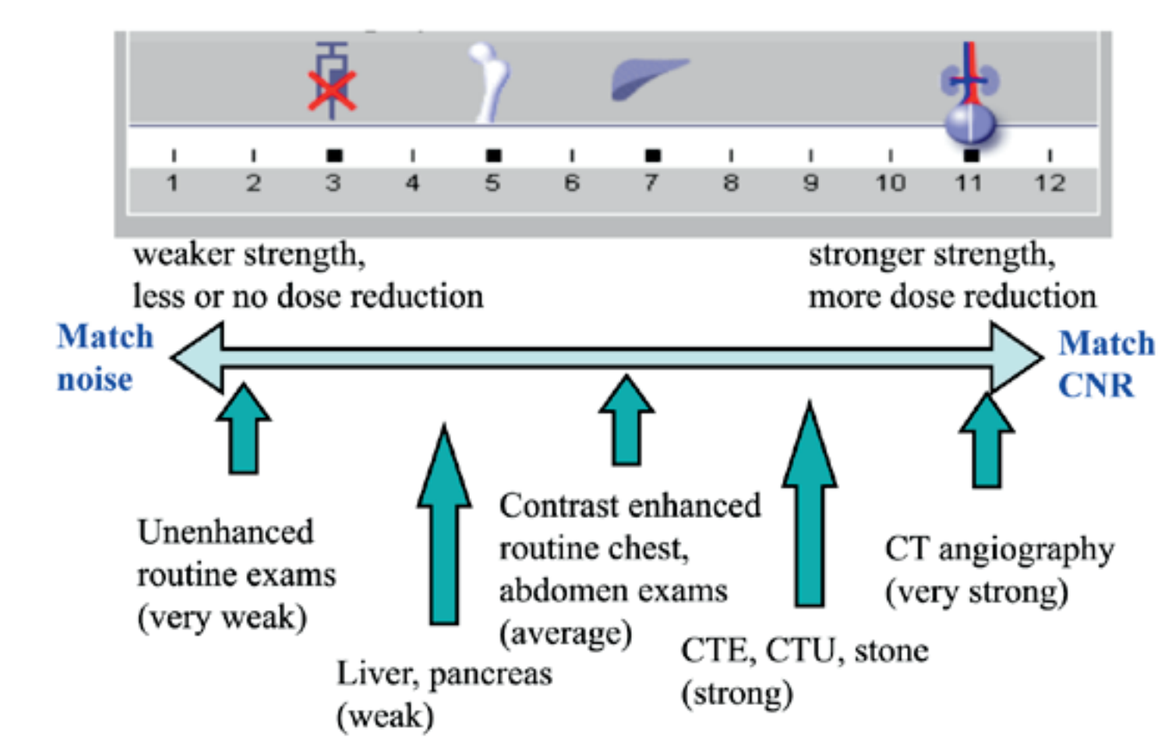
1 2 3 4 5 6 7 8 9 10 11 12
weaker strength, less or no dose reduction
stronger strength, more dose reduction
Match noise
Match iodine CNR
Unenhanced routine exams (very weak)
Liver, pancreas (weak)
Contrast enhanced routine chest, abdomen exams (average)
CTE, CTU, stone (strong)
CT angiography (very strong)

Figure 9. Different strength settings of an automatic kV selection tool for different diagnostic tasks. The slider bar position controls the strength setting, which corresponds to a contrast gain constraint parameter. With the change of the slider bar position, the strength can be anywhere between matching noise or iodine CNR. Towards the end of matching noise, strength is weaker and the chance of selecting a lower kV is lower with less or no dose reduction; Towards the end of matching iodine CNR, strength is stronger and the chance of selecting a lower kV is higher with more potential for dose reduction. Each diagnostic task may correspond to a certain level of strength setting, depending on the involvement of iodine contrast and the contrast level of the pathology of interest.

In a recent study with 101 CTA (CT angiography) exams (162 scans) and 91 contrast-enhanced abdomen-pelvis CT exams (113 scans) performed using the automatic kV selection tool, 80 or 100 kV was automatically used for 73% of the CTA scans and 54% of the abdomen-pelvis scans.(6,7) An overall radiation dose reduction of 29.6%±17.0% and 17%±15% compared with the reference 120 kV protocols were achieved for CTA scans and abdomen-pelvis scans, respectively (**Figure 10**). All exams were considered to have acceptable quality in terms of diagnostic confidence.

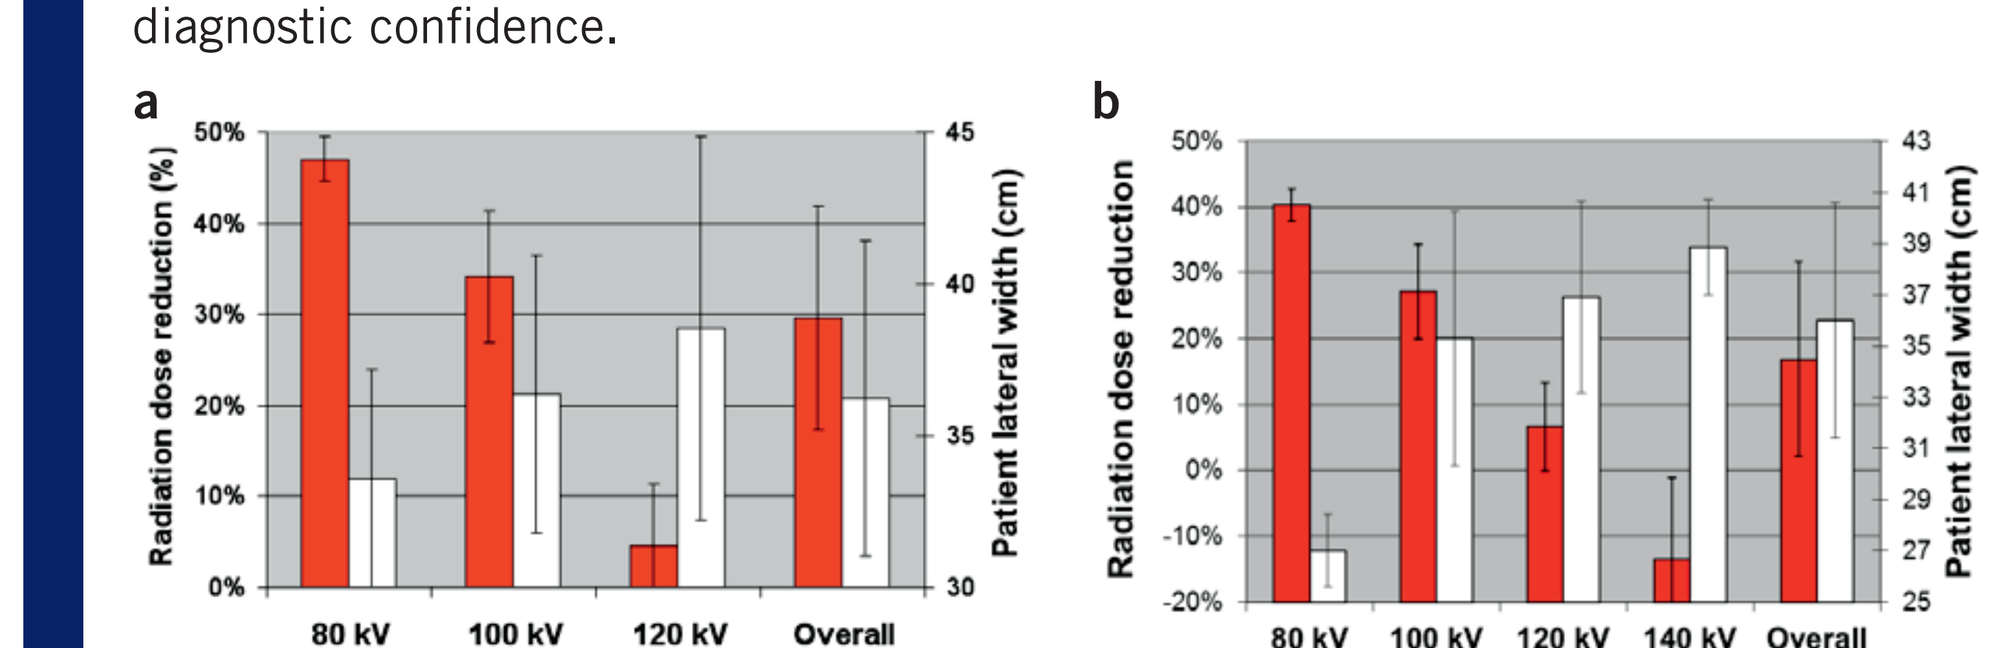
a
b
Radiation dose reduction (%)
Radiation dose reduction
Patient lateral width (cm)
80 kV
100 kV
120 kV
140 kV
Overall

Figure 10. Radiation dose reduction using an automatic kV selection tool in (a) 101 CTA exams and (b) 91 contrast-enhanced abdominal-pelvic CT exams. The mean and standard deviation of patient lateral width at each kV were also displayed. Red is dose reduction while white shows patient lateral width.

Important Considerations of Automatic kV Selection

Scan time and tube current limit

CT systems have a limit to the maximum tube current, and consequently the maximum radiation output. High scanning speed usually involves a fast rotation time and a high helical pitch, which limits the maximum radiation output, especially for lower kV. Therefore, when a fast scanning speed and a short scan time are desired, the lower kV may not be appropriate, even for a small-sized patient. It is essential to take into account the scan time and tube current limit in order to select the most appropriate kV.

Artifacts

There are two types of artifacts that tend to appear in scans acquired with lower kV. One is the photon starvation artifact caused by insufficient penetrating photons. In **Figure 1**, one can see that the image obtained with 80 kV for the 45 cm phantom contains much more severe photon starvation artifacts, suggesting that 80 kV should not be used at all for this patient size. The other type of artifact that could be of a potential concern for lower kV is streaking and dark shadow or banding artifacts when dense materials (e.g., highly concentrated iodine contrast media or metal) are present.

Other potential applications

Optimal kV can also be used for improving image quality or reducing the volume of iodine contrast used in the CT exam. For some challenging exams, one would rather generate the best possible image quality instead of reducing radiation dose in order to make a confident diagnosis on subtle pathologies. In this situation, one can use the optimal kV to maximize the image quality, with no need to reduce the radiation dose. For some patients with difficult intravenous accessibility or suboptimal renal function, one can also utilize the benefit of optimal kV to reduce the volume of iodine contrast injected to the patient.(8)

Clinical Examples

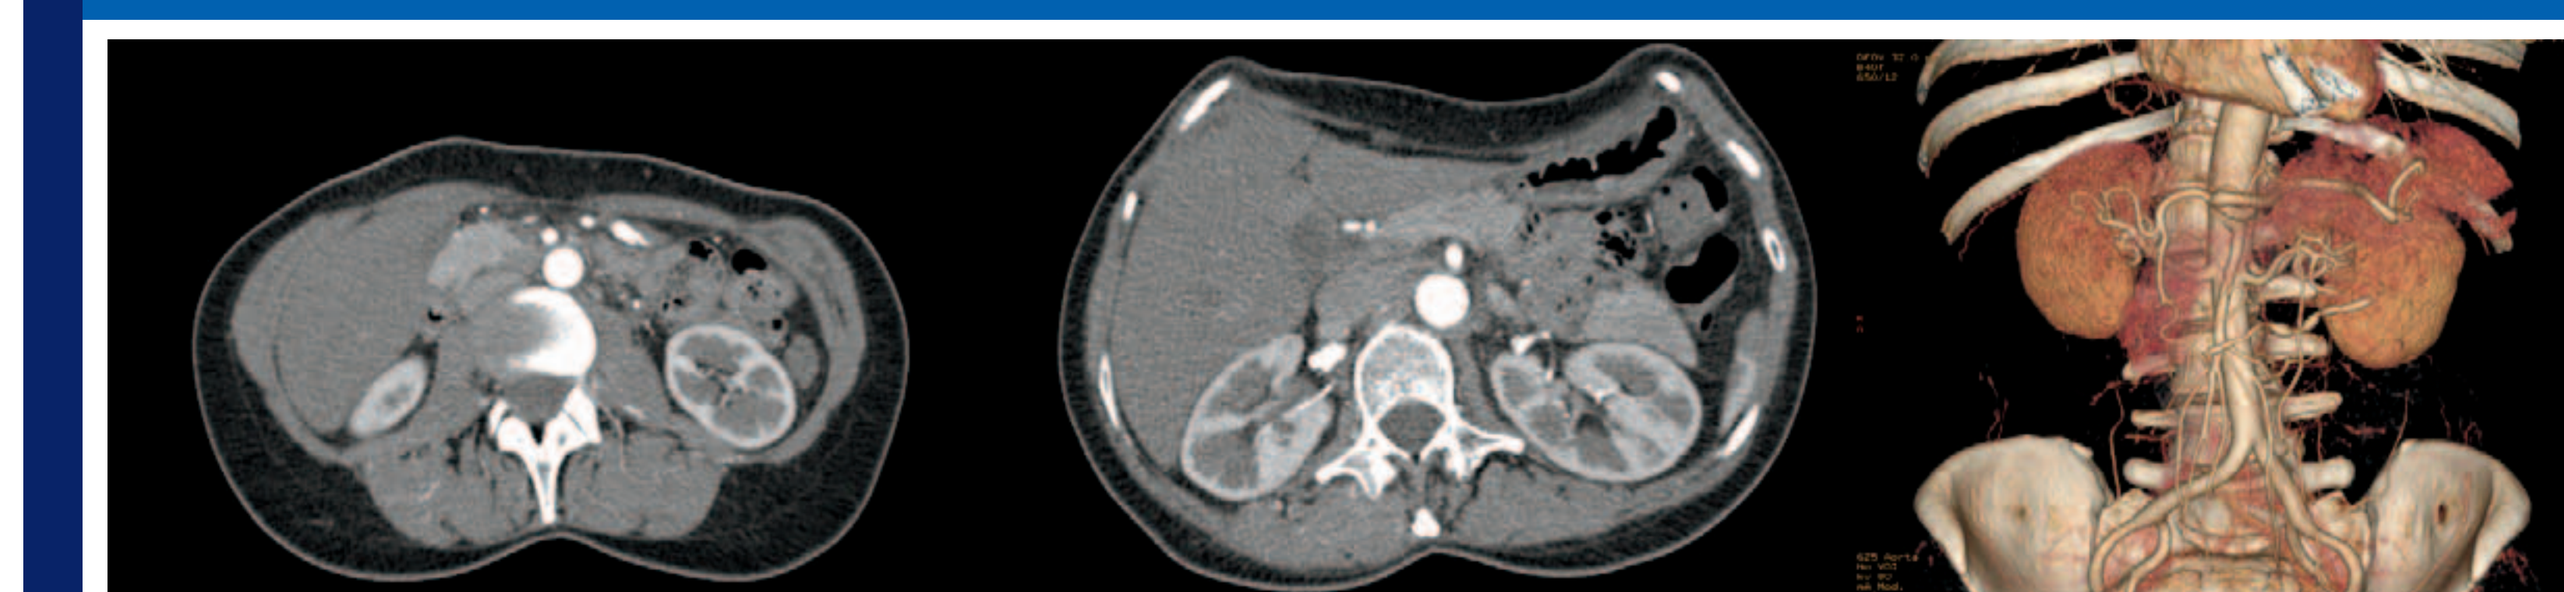

80 kV was automatically selected for this abdominal CTA exam (slider bar position 11). The patient had a relatively small size - 28 cm lateral width at the level of liver. A radiation dose reduction of 50% was achieved without sacrificing image quality. Scanning techniques: 80 kV, Quality reference mAs 467, effective mAs 291, CTDIvol 5.7 mGy.

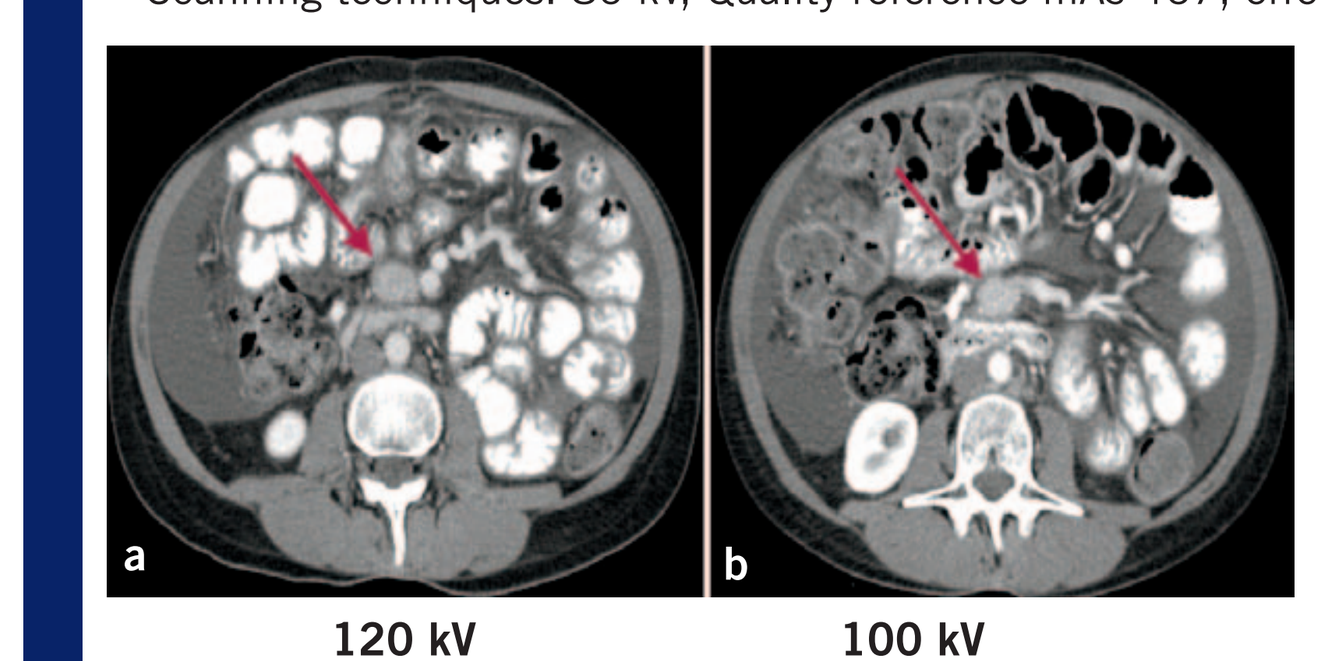
a
b
120 kV
100 kV

A 60 year old female with metastatic carcinoid to mesenteric root received two contrast-enhanced abdominal CT exams in a 6-month interval, one acquired with (a) CAREkV off, the other (b) with CAREkV on. 100 kV was automatically selected by the software with a 20 % dose reduction from the 120 kV prior scan (CTDIvol 17.0 → 13.5 mGy). There was no difference in image quality score.

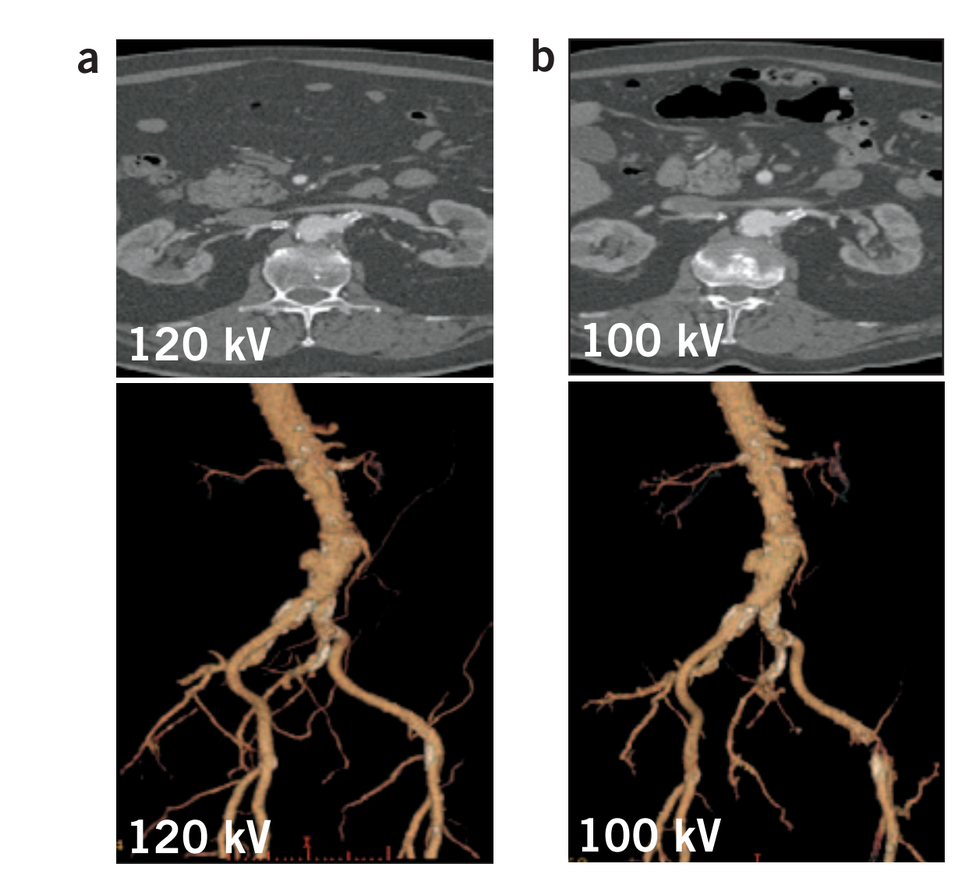
a
b
120 kV
100 kV
120 kV
100 kV

A 79 year old male with abdominal aortic aneurysm received two CT angiograms in a 1 year interval, one acquired with (a) CAREkV off, the other with (b) CAREkV on. 100 kV was selected by the software with a 31% dose reduction (from 28 mGy to 19.2 mGy). There was an improvement in image quality score. Compared with the reference 120 kV technique, a 40% dose reduction was achieved.

Conclusions

We described why the kV needs to be optimized and introduced the basic principles of automatic kV selection. We summarized recent development on automatic techniques to select the most dose-efficient kV. The appropriateness of using lower kV and dose reduction is dependent on patient size and diagnostic task, and is also affected by the system tube current limit and scanning speed. The use of lower kV should be carefully evaluated for each exam type in order to achieve an optimal tradeoff among contrast, noise, artifacts, and scanning speed.

References

1. McCollough CH, Bruesewitz MR, Kofler JM, Jr. CT dose reduction and dose management tools: overview of available options. Radiographics 2006; 26:503-512.
2. Guimaraes LS, Fletcher JG, Harmsen WS, et al. Appropriate Patient Selection at Abdominal Dual Energy CT: Relationship between Patient Size, Image Noise and Image Quality. Radiology 2010 (In Press).
3. McCollough CH. Automatic exposure control in CT: are we done yet? Radiology 2005; 237:755-756.
4. Yu L, Bruesewitz MR, Thomas KB, Fletcher JG, Kofler JM, McCollough CH. Optimal tube potential for radiation dose reduction in pediatric CT: principles, clinical implementations, and pitfalls. Radiographics 2011; 31:835-848.
5. Yu L, Li H, Fletcher JG, McCollough CH. Automatic selection of tube potential for radiation dose reduction in CT: A general strategy. Medical Physics 2010; 37:234-243.
6. Yu L, Fletcher J, Grant K, et al. Automatic kV Selection for Radiation Dose Reduction in Contrast-enhanced Abdominal CT. In:97th Scientific Assembly and Meeting of the Radiological Society of North America. Chicago, IL, 2011.
7. Yu L, Fletcher J, Vrtiska T, et al. Automatic kV Selection for Radiation Dose Reduction in CT Angiography. In:97th Scientific Assembly and Meeting of the Radiological Society of North America. Chicago, IL, 2011.
8. Hough D, Yu L, Shiung M, et al. The Lymphoma Follow-up CT: Age-appropriate Individualization to Decrease IV Contrast Dose or Radiation Dose. In:97th Scientific Assembly and Meeting of the Radiological Society of North America. Chicago, IL, 2011.

© 2011 Mayo Foundation for Medical Education and Research